A prospective, single-arm, phase I/II trial of RAS peptide vaccine TG01/GM-CSF and gemcitabine as adjuvant therapy for patients with resected pancreatic adenocarcinoma

Daniel H. Palmer[1], Svein Dueland[2], Juan W. Valle[3], Tone Otterhaug[4], Jon Amund Eriksen[4], Helene Muller[5], Berit Iversen[4], Robert M. Miller[6];

[1]University of Liverpool Cancer Research UK Centre, Liverpool, United Kingdom; [2]The Norwegian Radium Hospital, Oslo, Norway; [3]Institute of Cancer Studies, University of Manchester, The Christie Hospital, Manchester, United Kingdom; [4]Targovax AS, Lysaker, Norway; [5]Artemida Pharma Ltd, Strasbourg, France; [6]Artemida Pharma Ltd, Stevenage, United Kingdom

Background

TG01 is a cancer specific therapeutic peptide vaccine targeting oncogenic mutations in the RAS genes (K-, N- and H-RAS). RAS mutation is a driver for development of cancer and is present in up to 30% of all cancer[1] and above 85% in pancreatic cancer[2]. There is a large unmet medical need for treatment of cancers with RAS mutation. TG01 induces RAS mutation specific T cell responses.

TG01 consists of a mixture of 7 synthetic peptides that represent 7 of the most common codon 12 and 13 mutations in p21RAS associated with human cancer. Molgramostim (recombinant human granulocyte macrophage-colony stimulating factor (GM-CSF)) is co-administered with TG01 in order to enhance induction of the T cell responses.

TG01 is the first therapeutic peptide vaccine targeting RAS that entered clinical trials.
Earlier studies demonstrate that adjuvant vaccination with TG01/GM-CSF given as monotherapy to pancreatic cancer patients after tumor resection induce immune response in 100% of evaluable patients[3].

Adjuvant treatment of resected pancreatic cancer with gemcitabine are now widely used and the present study evaluates safety and immunological response of the RAS peptide vaccine TG01/GM-CSF when given in combination with gemcitabine as adjuvant therapy.

Methods

Eligible patients had a confirmed diagnosis of adenocarcinoma of the pancreas and successful surgical resection (R0 or R1), no previous radiation or chemotherapy (except for primary neoadjuvant chemotherapy, if applicable) and were expected to receive gemcitabine as adjuvant chemotherapy within 12 weeks of surgery.

Patients started TG01/GM-CSF within 1-8 weeks after surgery and chemotherapy with Gemcitabine (1000 mg/m² for 3/4 weeks x 6 cycles) 3-7 weeks after vaccination or started TG01/GM-CSF and chemotherapy concomitantly within 12 weeks of surgery, (Figure 1).

The patients were assessed for immune responses (DTH) up to week 11 after receiving at least one cycle of gemcitabine. Safety information was collected during the entire 2-year study period. Patients who could not tolerate gemcitabine could be switched to 5-FU/leucovorin.

Figure 1. TG01-01 Study Design

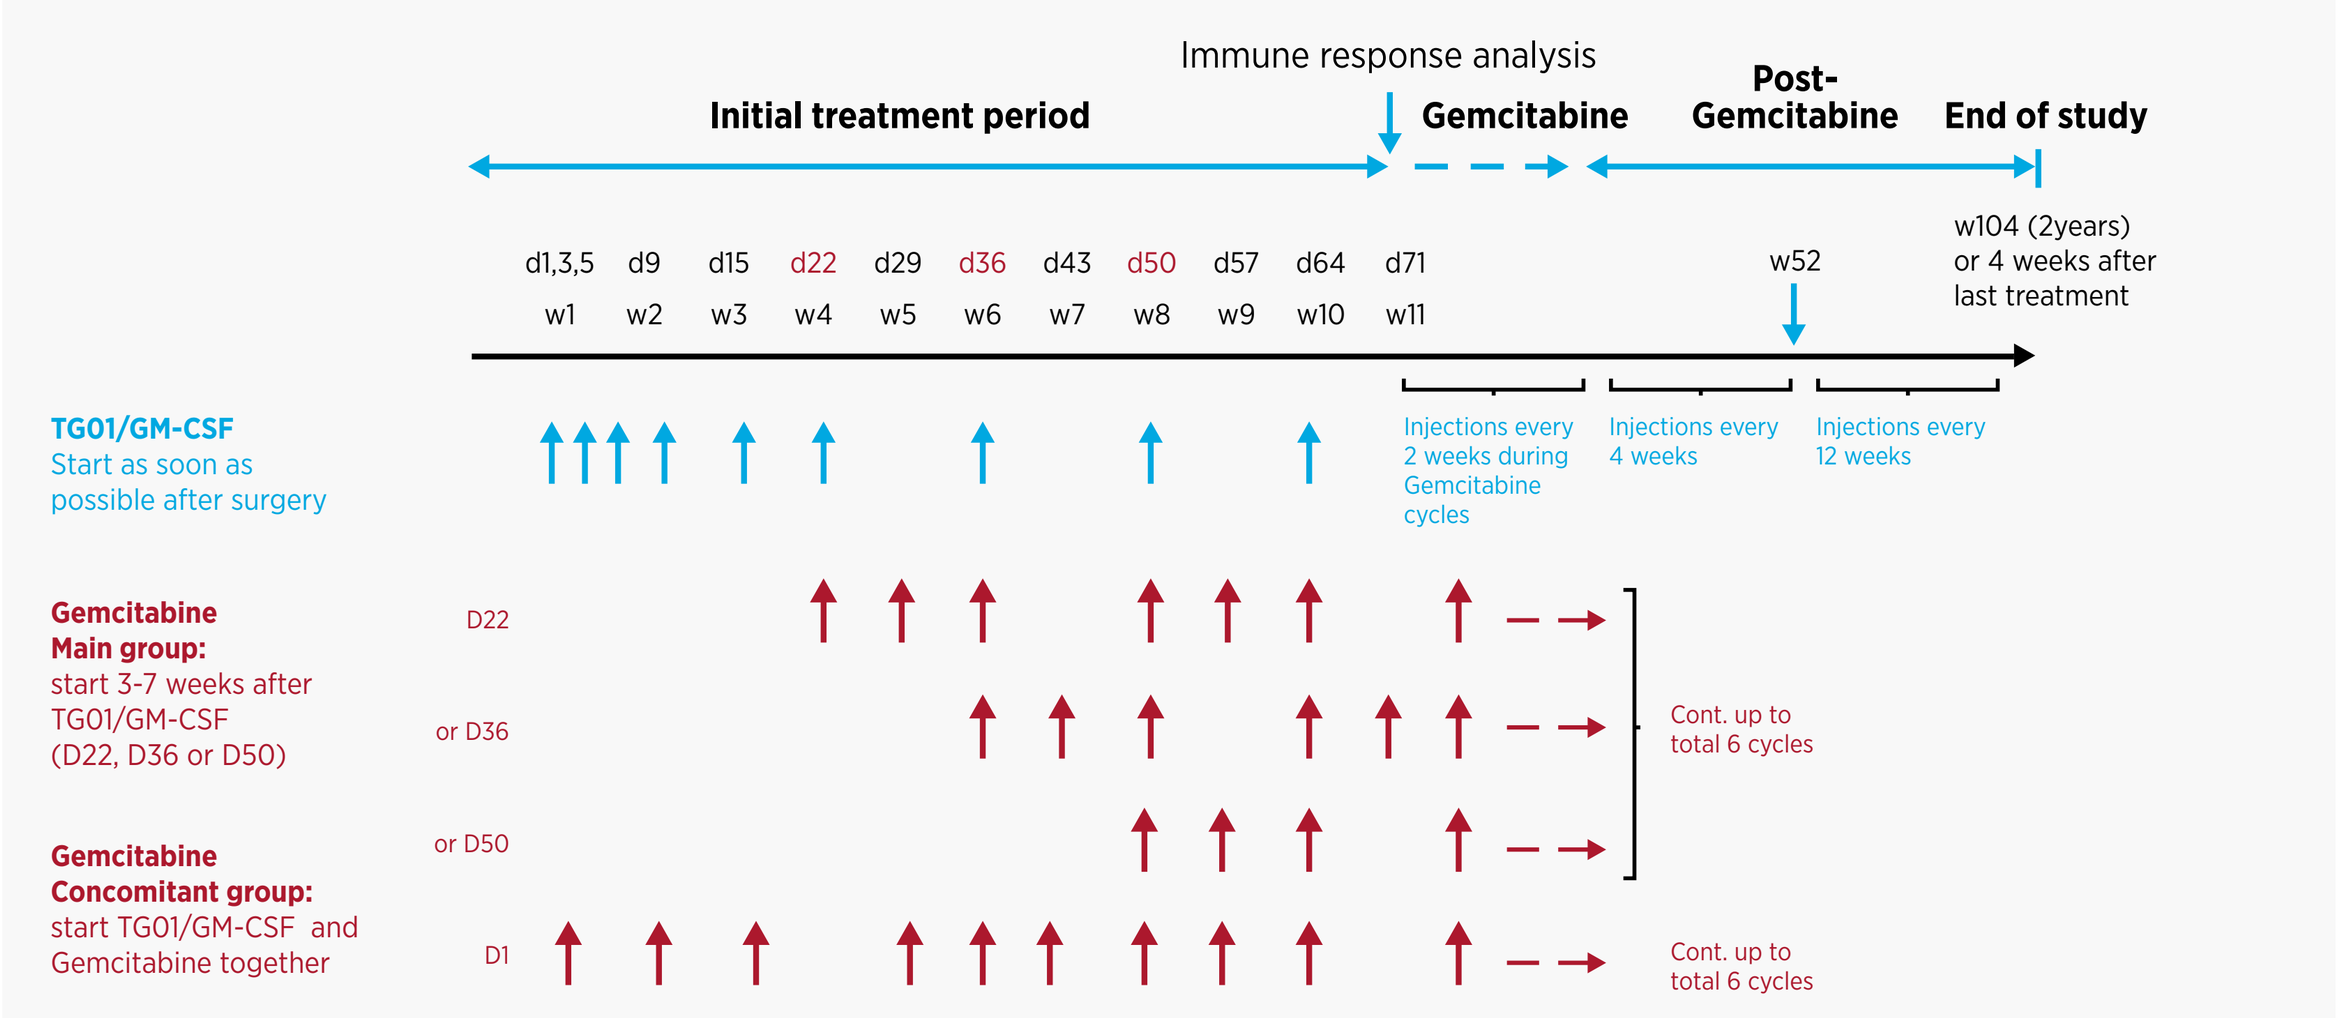
Immune response analysis
Initial treatment period
Gemcitabine
Post-Gemcitabine
End of study
d1,3,5 d9 d15 d22 d29 d36 d43 d50 d57 d64 d71
w1 w2 w3 w4 w5 w6 w7 w8 w9 w10 w11
w52
w104 (2years) or 4 weeks after last treatment
TG01/GM-CSF
Start as soon as possible after surgery
Injections every 2 weeks during Gemcitabine cycles
Injections every 4 weeks
Injections every 12 weeks
Gemcitabine
Main group:
start 3-7 weeks after TG01/GM-CSF
(D22, D36 or D50)
D22
or D36
or D50
Gemcitabine
Concomitant group:
start TG01/GM-CSF and Gemcitabine together
D1
Cont. up to total 6 cycles
Cont. up to total 6 cycles

Results

This is an ongoing study and preliminary data as of 25th April 2015 are presented. 19 patients have been included from three sites (1 in Norway and 2 in UK), 18 patients were eligible for immune assessment in initial treatment period (up to week 11). Patient baseline characteristics are shown in table 1.

Table 1. Baseline Characteristics

Parameter	N = 19
Age (y), median (range)	67 (49, 79)
Gender, n (%)	
Male	10 (53%)
Female	9 (47%)
Baseline ECOG score, n (%)	
0	8 (42%)
1	11 (58%)
Outcome surgery, n (%)	
R0	6 (32%)
R1	13 (68%)

Four patients are still ongoing (3-6 months after the start of vaccinations) and 15 patients have discontinued from the study (Table 2). Six patients had confirmed disease recurrence (1 died due to disease progression).

Table 2. Patient Disposition

	Number of Patients (N=19)
Patienst ongoing	4
Disease Recurrence (1 death from progression)	6
Withdrawals:	
Consent withdrawn	3
Investigators decision	2
Adverse events (including 1 unrelated death)	4

Immunological response

Overall immune response as detected by delayed type hypersensitivity (DTH) testing in the skin and in vitro T cell proliferation are presented in Table 3. Positive DTH response (average diameter ≥5mm) was detected in 14/18 (78%) treated patients, 14/17 (82%) evaluable patients, 13/14 (93%) in main group and 1/3 (33%) in concomitant group.

Table 3. Immune responses detected by DTH and in vitro T cell proliferation

	Positive DTH response n=17*	Positive T cell response n=8**
Total	14/17 (82%)	6/8 (75%)***
Main group	13/14 (93%)	6/8 (75%)***
Concomitant Group	1/3 (33%)	0/0

*18 patients treated, 17 patients evaluable for DTH response

**10 patients analyzed per to date for T cell response, 8 patients evaluable for assessment

*** 2 patients had SI ≥1.5 and <2

The KRAS mutation status and detected immune responses for individual patients are summarized in Table 4.

KRAS mutation (codon 12/13) were detected in 15/17 (88%) of the analyzed patients. 14/17 (82%) patients had developed detectable DTH skin reaction to TG01 by week 11, all being positive at week 6 or week 8. 1/3 (33%) evaluable patients in the concomitant group had detectable DTH.

Possible causes for the observed drop in DTH responses after second cycle of chemo (week 10) might be the effects of prolonged chemotherapy and/or natural fluctuations[4].

In vitro T cell analyses showed induction of TG01 specific immune responses (Stimulation index (SI) ≥2) in 6/8 (75%) evaluable patients analyzed to date, all in main group. As shown in Figure 2a, the immune response increased over time after continuous vaccinations. Immune responses were detected for all single peptides covered in TG01 demonstrating that all peptides are immunogenic (Figure 2b).

Table 4. Detected KRAS codon 12 or 13 mutation and immune response

a. Main group; TG01/GM-CSF from week 1, TG01/GM-CSF and gemcitabine from week 4

Main group (n=14)	KRAS mutation	DTH W1 (D1)	DTH W1 (D8)	DTH W3 (D15)	DTH W4 (D22)	DTH W6 (D36)	DTH W8 (D50)	DTH W10 (D64)	DTH Outcome	In vitro T cell response W11
Pt 01-001	yes	-	-	-	-	+	+	-/+	+	+
Pt 01-002	yes	-	-	-	-	-	+	-	+	+
Pt 01-003	yes	-	-	-	-	+	+	+	+	-/+
Pt 01-005	yes	-	-	+	-/+	+	+	-	+	-/+
Pt 01-006	yes	-	-	+	+	+	+	+	+	+
Pt 01-007	yes	-	-	-	-	-	+	-	+	NE
01-008	yes	-	+	+	+	+	+	+	+	+
Pt 01-009	yes	-	-	+	+	+	+	-	+	+
Pt 01-010	yes	-	-	-	+	+	+	+	+	+
Pt 01-011	no	-	-	+	+	-	+	+	+	NE
Pt 02-001*	no	-	-	-	-	-	-	NC	-	NC
Pt 02-002	yes	-	-	-	+	+	+	+	+	NT
Pt 02-003	yes	-	-	+	+	+	-	-	+	NT
Pt 02-004	yes	-	-	+	+	NC	NC	NC	+	NT

b. Concomitant group; TG01/GM-CSF and gemcitabine from week 1

Concomitant group (n=14)	KRAS mutation	DTH W1 (D1)	DTH W1 (D8)	DTH W3 (D15)	DTH W5 (D29)	DTH W7 (D43)	DTH W9 (D57)	DTH W11 (D71)	DTH Outcome	In vitro T cell response W11
Pt 02-005	yes	-	-	-	-	-	-	-	-	NT
Pt 02-006	yes	-	-	-	NC	NC	NC	NC	NE	NT
Pt 02-007	yes	-	-	+	-	+	+	+	+	NT
Pt 03-001	NT	NC	-	-	-	-	-	-	-	NT

DTH detection: + : Average diameter ≥ 5mm, - : ≥0 and <2, -/+ : Average diameter ≥2 and <5
T cell response: +: SI ≥2, -/+: SI ≥1.5 and <2
NE: Not Evaluable, NC: Sample not collected, NT: Not Tested
*DTH responses only assessed up to week 8

Figure 2. In vitro T cell immune responses to TG01 and single peptides

a. Immune response to TG01 increases over time after continous TG01 vaccination

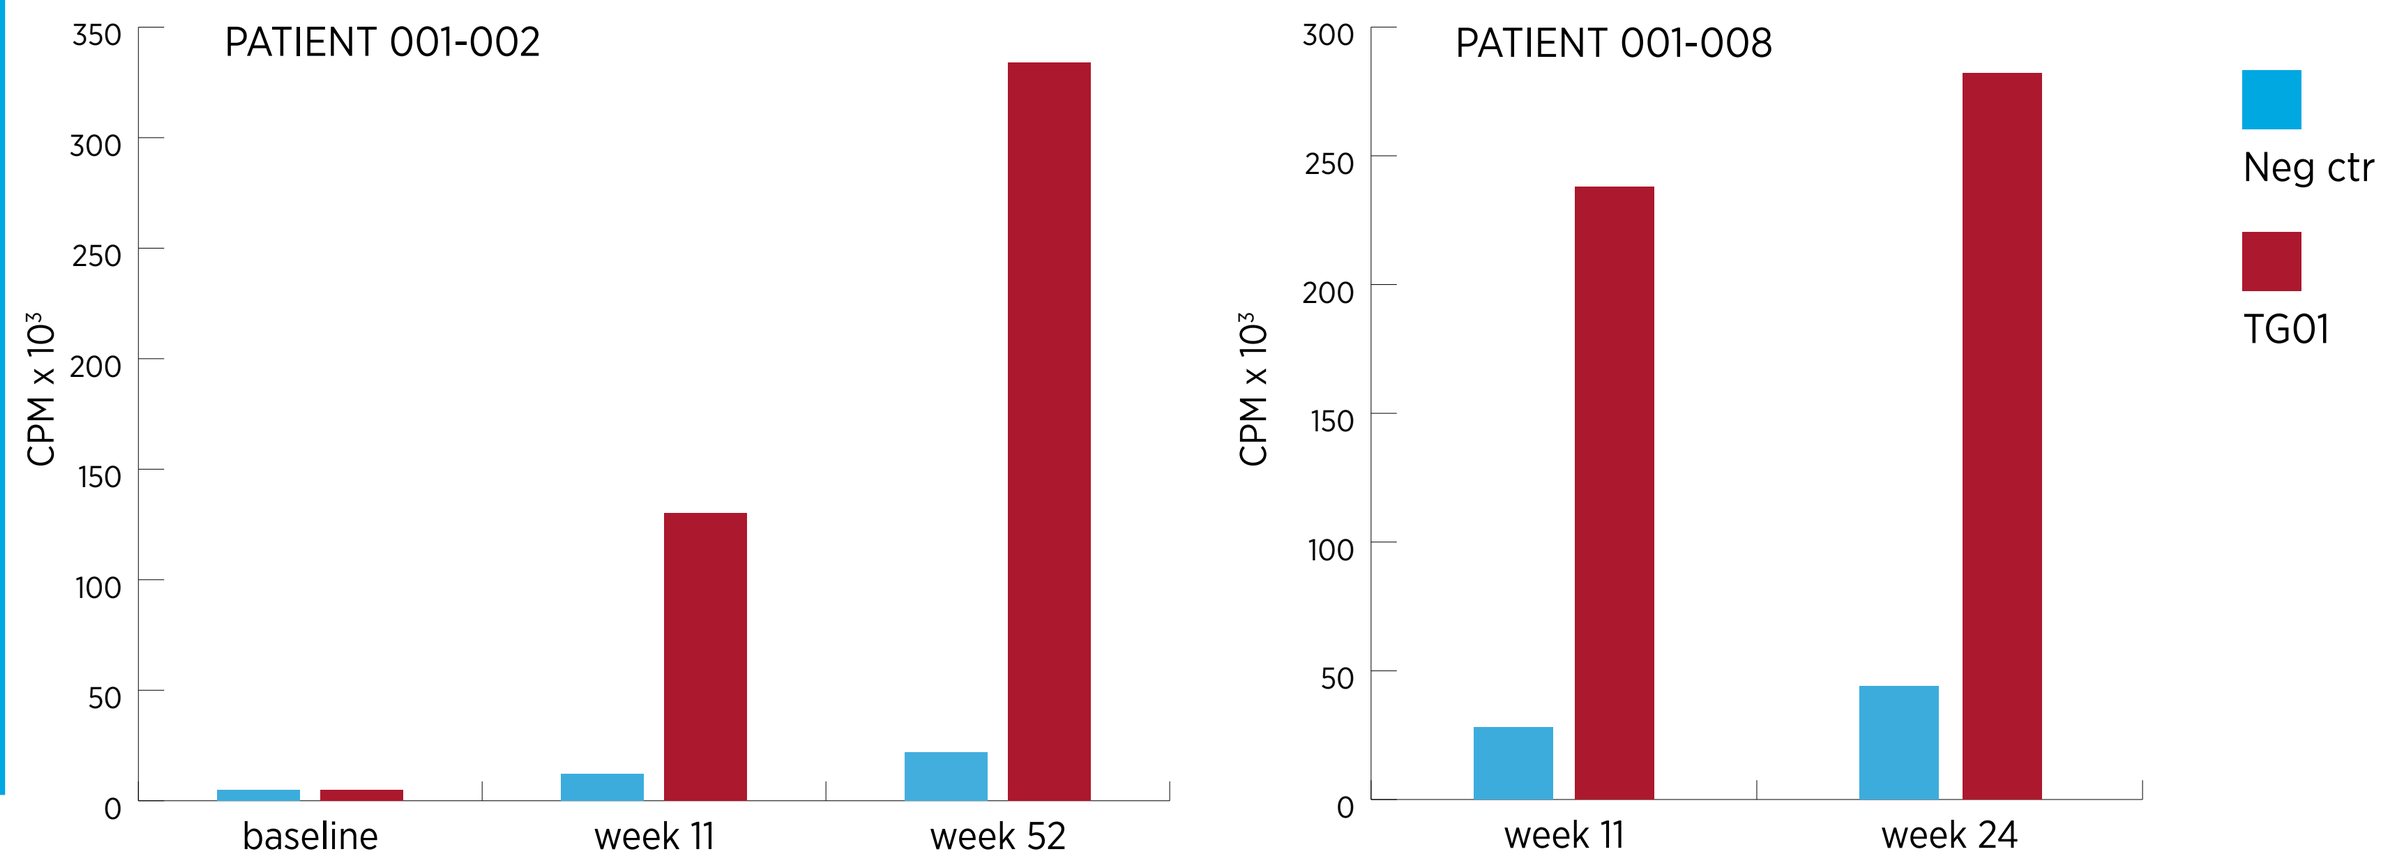
PATIENT 001-002
PATIENT 001-008
CPM x 10³
baseline
week 11
week 52
week 11
week 24
Neg ctr
TG01

b. Immune response was detected towards all 7 peptides that comprises TG01

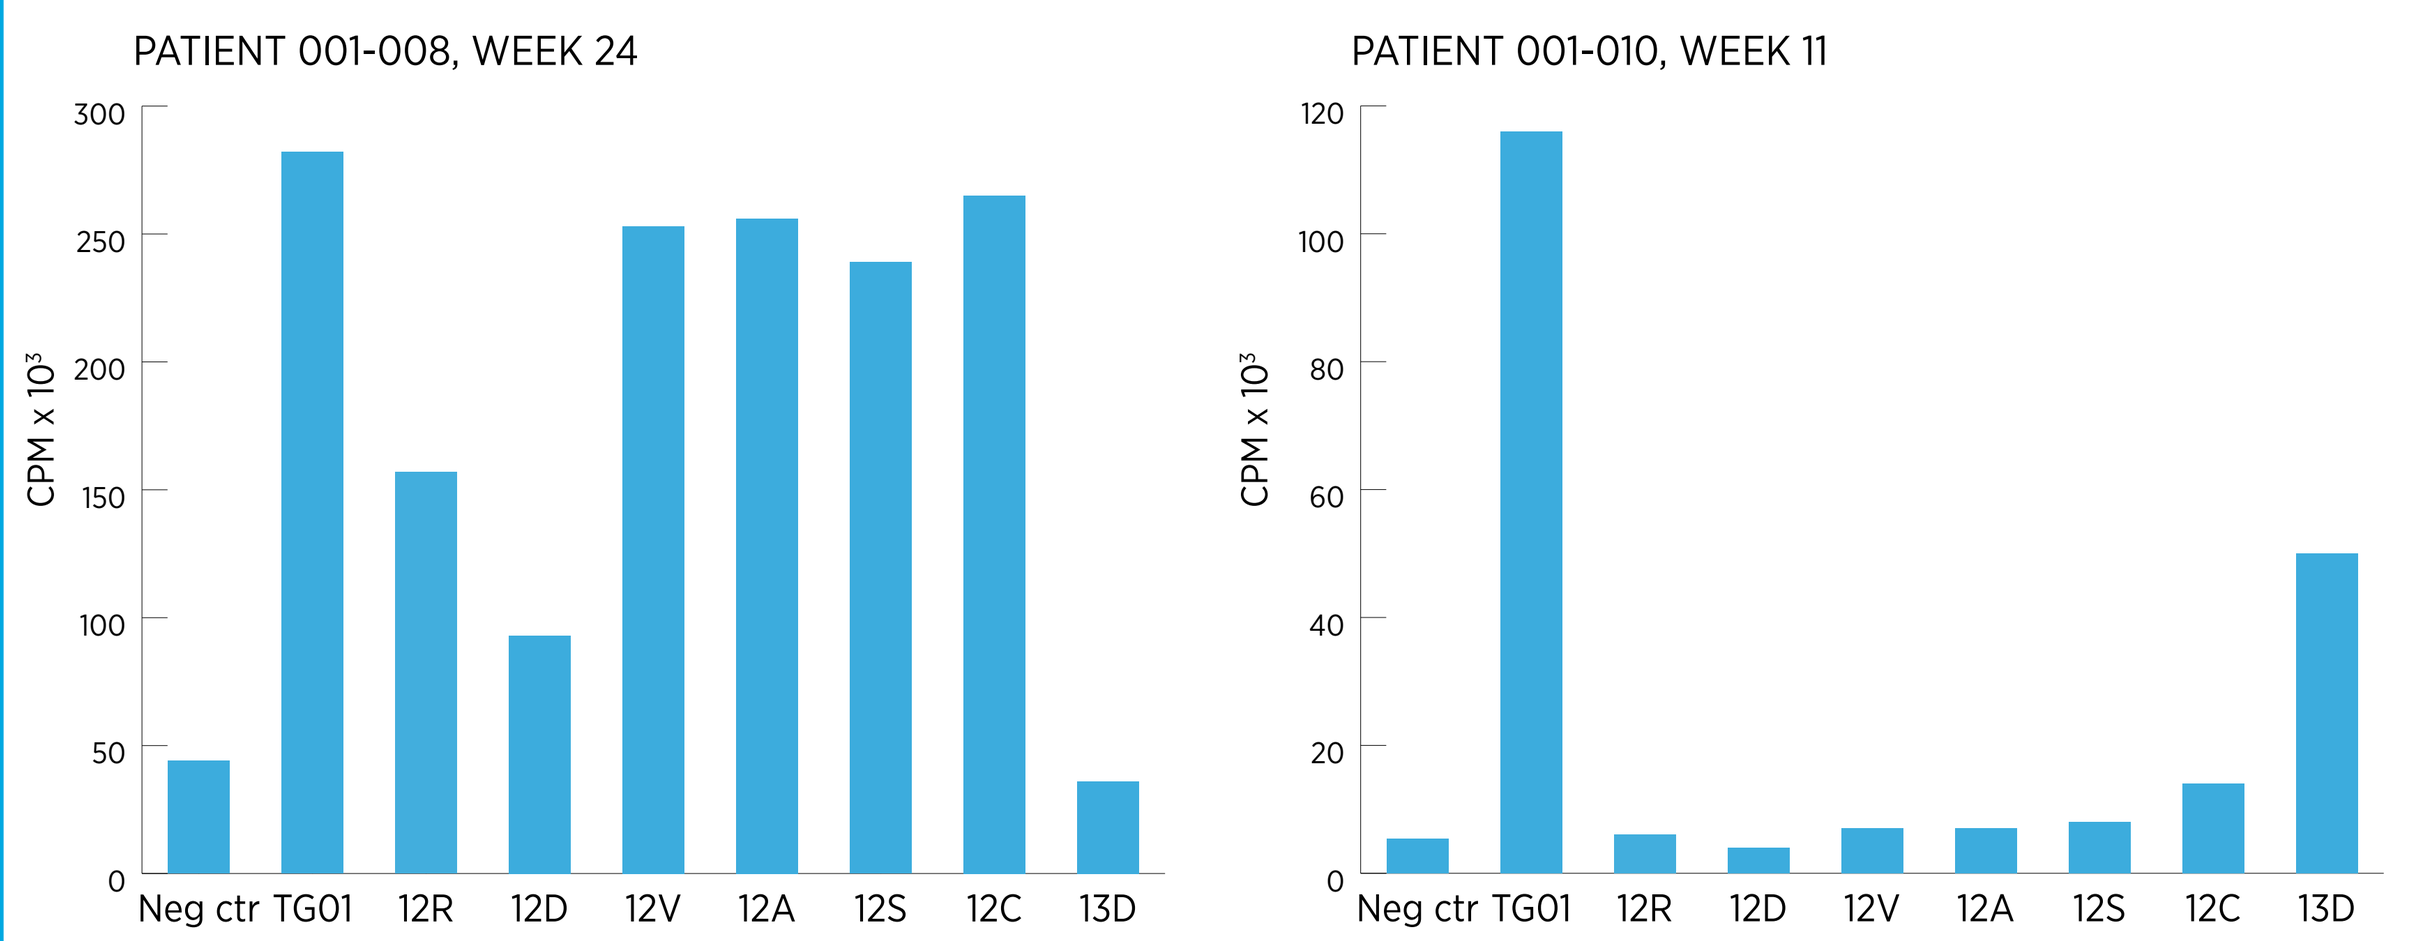
PATIENT 001-008, WEEK 24
PATIENT 001-010, WEEK 11
CPM x 10³
Neg ctr TG01 12R 12D 12V 12A 12S 12C 13D

Safety

So far 251 adverse events (AEs) have been reported in 19 patients (Table 5).
The most common AEs reported (occurring in at least 3 patients) are presented in table 5.
The number of drug related grade 3/4 events are included in table 6.

Table 5. Adverse events (all cause) occuring in at least 3 patients, number of grade 3/4 events and number of patients with grade 3/4 events

System organ class Preferred term	Events	Patients (N=19) n (%)	Grade 3/4 events	Patients with Grade 3/4 events n (%)
Adverse event				
Any adverse event	251	19	35	12 (68%)
Blood and lymphatic system disorders				
Neutropenia (including neutrophil count decrease)	13	8 (42%)	13	8 (42%)
Gastrointestinal disorders				
Nausea	33	9 (47%)		
Abdominal pain	12	8 (42%)	2	2 (11%)
Diarrhea	5	5 (26%)	1	1 (5%)
Vomiting	14	5 (26%)		
Constipation	3	3 (16%)		
General disorders and administration site conditions				
Fatigue	16	10 (53%)	1	1 (5%)
Influenza-like illness	10	6 (32%)		
Injection Site Reaction	8	4 (21%)		
Pyrexia (fever)	7	4 (21%)		
Edema peripheral	3	3 (16%)		
Metabolism and nutrition disorders				
Decreased appetite	5	5 (26%)		
Hyperglycemia	3	3 (16%)	3	3 (16%)
Immune system disorders				
Anaphylaxis	3	3 (16%)	3	3 (16%)
Nervous system disorders				
Headache	6	4 (21%)		
Dysgeusia	3	3 (16%)		
Psychiatric disorders				
Insomnia	4	4 (21%)		

32 of the 251 AEs reported so far, have been related to the vaccination; 29 related to TG01/GM-CSF, 3 related to TG01 and 1 related to GM-CSF. The majority of these adverse reactions were expected skin reactions as well as flu-like symptoms. Most of these events were grade 1/2, except for the two anaphylactic reactions reported as grade 4. Four patients experienced allergic reactions related to TG01 or TG01/GM-CSF. Two experienced a serious adverse event of anaphylaxis, one patient experienced symptoms of an allergic reaction on 2 occasions and the last patient experienced dizziness, lightheaded, puffiness of both eyes. The more severe allergic reactions were those that occurred after gemcitabine had been administered for at least 3 cycles.

Table 6. Grade 3/4 Adverse Events related to any study treatments

Preferred term	Grade ≥3 events	Relationship to study medication: TG01/GM-CSF	TG01	GM-CSF	Gemcitabine/5-FU + Leucovorin
Neutropenia	13				13
Anemia	1				1
Anaphylactic	2	1	1		
Hemoglobin Decrease	1				1
Platelet count decrease	1				1
Hypertension	1				1

Table 7. Serious Adverse Events

Serious Adverse Events Preferred term	Number of Events	Relationship to study treatment
Anaphylactic reaction	2	Related to TG01 +/- GM-CSF
Dyspnea	1	Related to Gemcitabine and TG01/GM-CSF
Pulmonary infection	1	Related to Gemcitabine
Pyrexia (fever)	2	
Anemia	1	
Anaphylactic shock related to a concomitant medication (Emend)	1	Unrelated to study treatments
Hyperglycemia	1	
Urosepsis	1	
Pneumonia	1	

11 serious adverse events occurred in six patients including 1 unrelated death. Three of these events are considered to be related to TG01/GM-CSF (Table 7).

Conclusions

- Persisting RAS mutation specific T cell responses were induced and enhanced when TG01/GM-CSF was administered in combination with gemcitabine.
- The T cell responses were maintained during and after chemotherapy with booster vaccinations.
- The preliminary immunological results indicate that initiating the TG01/GM-CSF treatment prior (3 weeks) to start of gemcitabine treatment might be favorable for induction of immune response – at least as measured by DTH.
- The regimen was generally well tolerated with related events to TG01/GM-CSF being those expected (local reactions and flu-like symptoms) for a peptide vaccine. Grade 3/4 reactions were primarily related to gemcitabine. There were 4 patients with related allergic reactions to vaccination (6 events) for three of which the event occurred during or after gemcitabine treatment and in 2 cases it was severe. Additional vaccination regimens are currently under investigation.

References

[1] Alberto Fernández-Medarde and Eugenio Santos; Genes and Cancer: 2(3), 344-358 (2011)
[2] Miglio U et al. Pathol Res Pract; 210(5):307-11 (2014)
[3] Weden S et al. Int. J. Cancer: 128; 1120-1128 (2011)
[4] Gjertsen MK et al. Int. J. Cancer: 92, 441-450 (2001)

www.targovax.com